GPV381: A first-in-class therapeutic vaccine neutralizing gingipain toxins that drive chronic inflammation

Paul Cockle[1], Sara Hadjigol[2], James Holden[2], Allister Loughran[1], Neil O'Brien-Simpson[2], Michal Pasternak[1], Eric Reynolds[2], William Singleton[2], Lori Sun[1], Sean McLoughlin[1]
(1) Denteric, Melbourne, VIC, Australia; (2) Melbourne Dental School, University of Melbourne, Melbourne, VIC, Australia

Pg drives severe disease through gingipain toxins

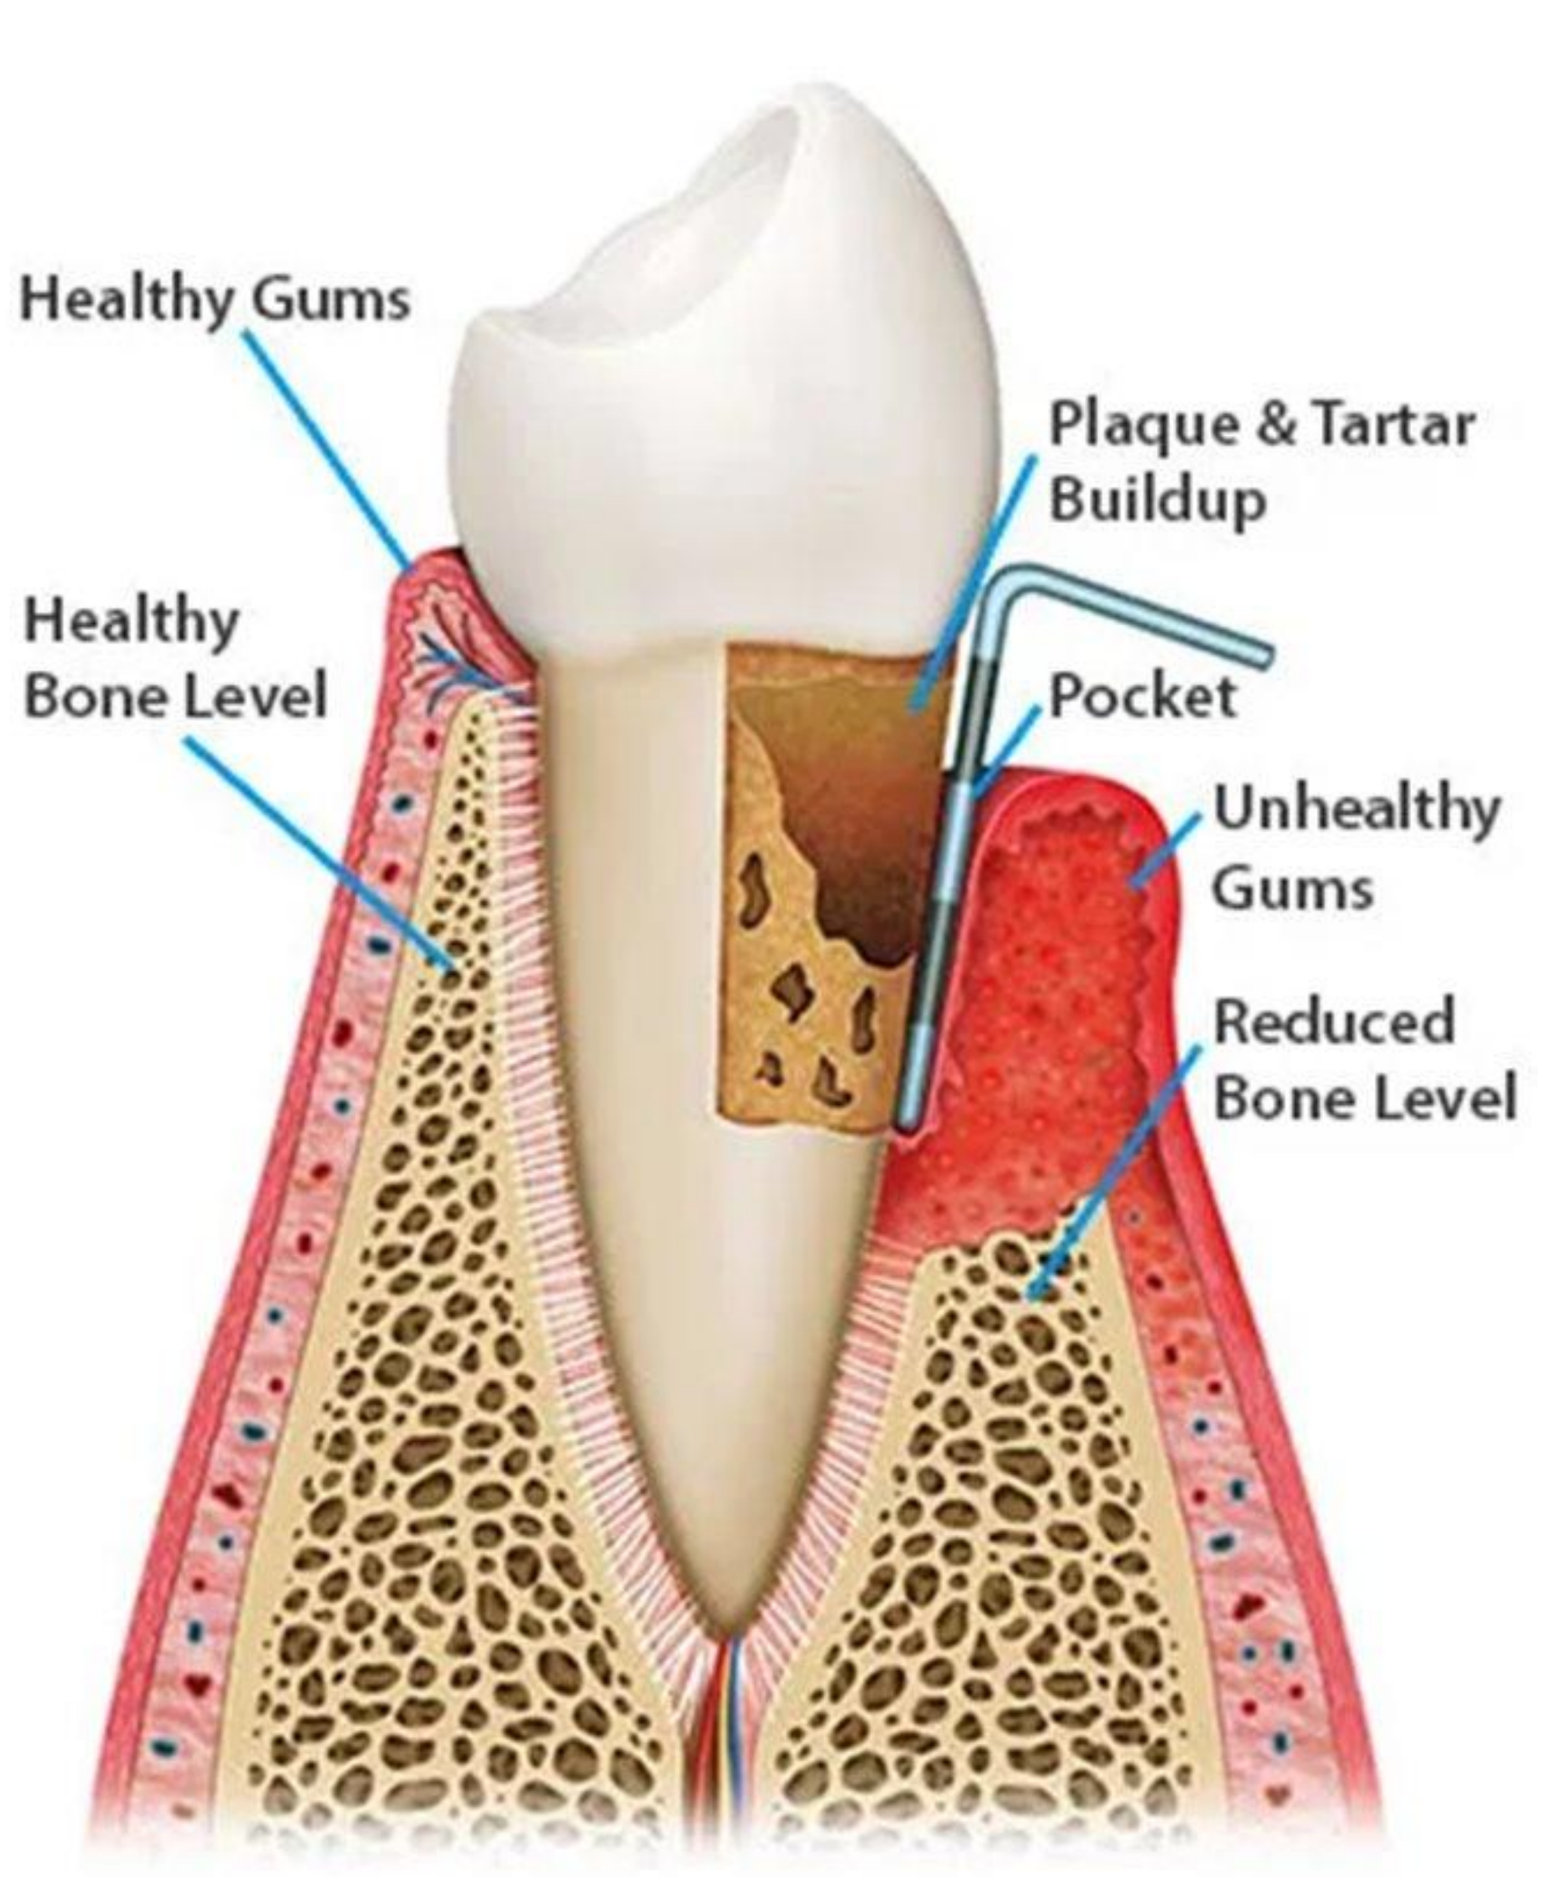
Healthy Gums
Healthy Bone Level
Plaque & Tartar Buildup
Pocket
Unhealthy Gums
Reduced Bone Level

- ***Porphyromonas gingivalis (Pg)*** is an anaerobic bacteria and its levels increase with pocket depth
- Higher Pg levels correlate with **increasing disease severity** and bone loss
- While many bacteria are present in periodontitis, only Pg produces **gingipain toxins**, the central drivers of **immune dysfunction** and **chronic inflammation**.
- Current SoC is Scaling and Root Planing (**SRP**), a mechanic process that removes plaque, which only provide **short term benefits**

Scientific rationale behind GPV381

Gingipain toxins drive immune dysfunction

- Gingipain toxins are upstream drivers of chronic inflammation, keeping inflammatory pathways activated in immune cells while impairing effective bacterial clearance

Neutralizing gingipains restores immune control

- Neutralizing gingipain toxins restores immune control, allowing inflammation to resolve and bacterial clearance to resume

GPV381 induces gingipain neutralizing antibodies

- GPV381 generates durable neutralizing antibodies against gingipain toxins
- Phase 2 is designed to demonstrate clinically meaningful disease modification, including reduction in inflammation and healing in periodontal pockets
- Delivering this would shift treatment guidelines and redefine how periodontitis is managed

GPV381-induced antibodies neutralize the two critical domains of gingipain toxins

Adhesin inhibition

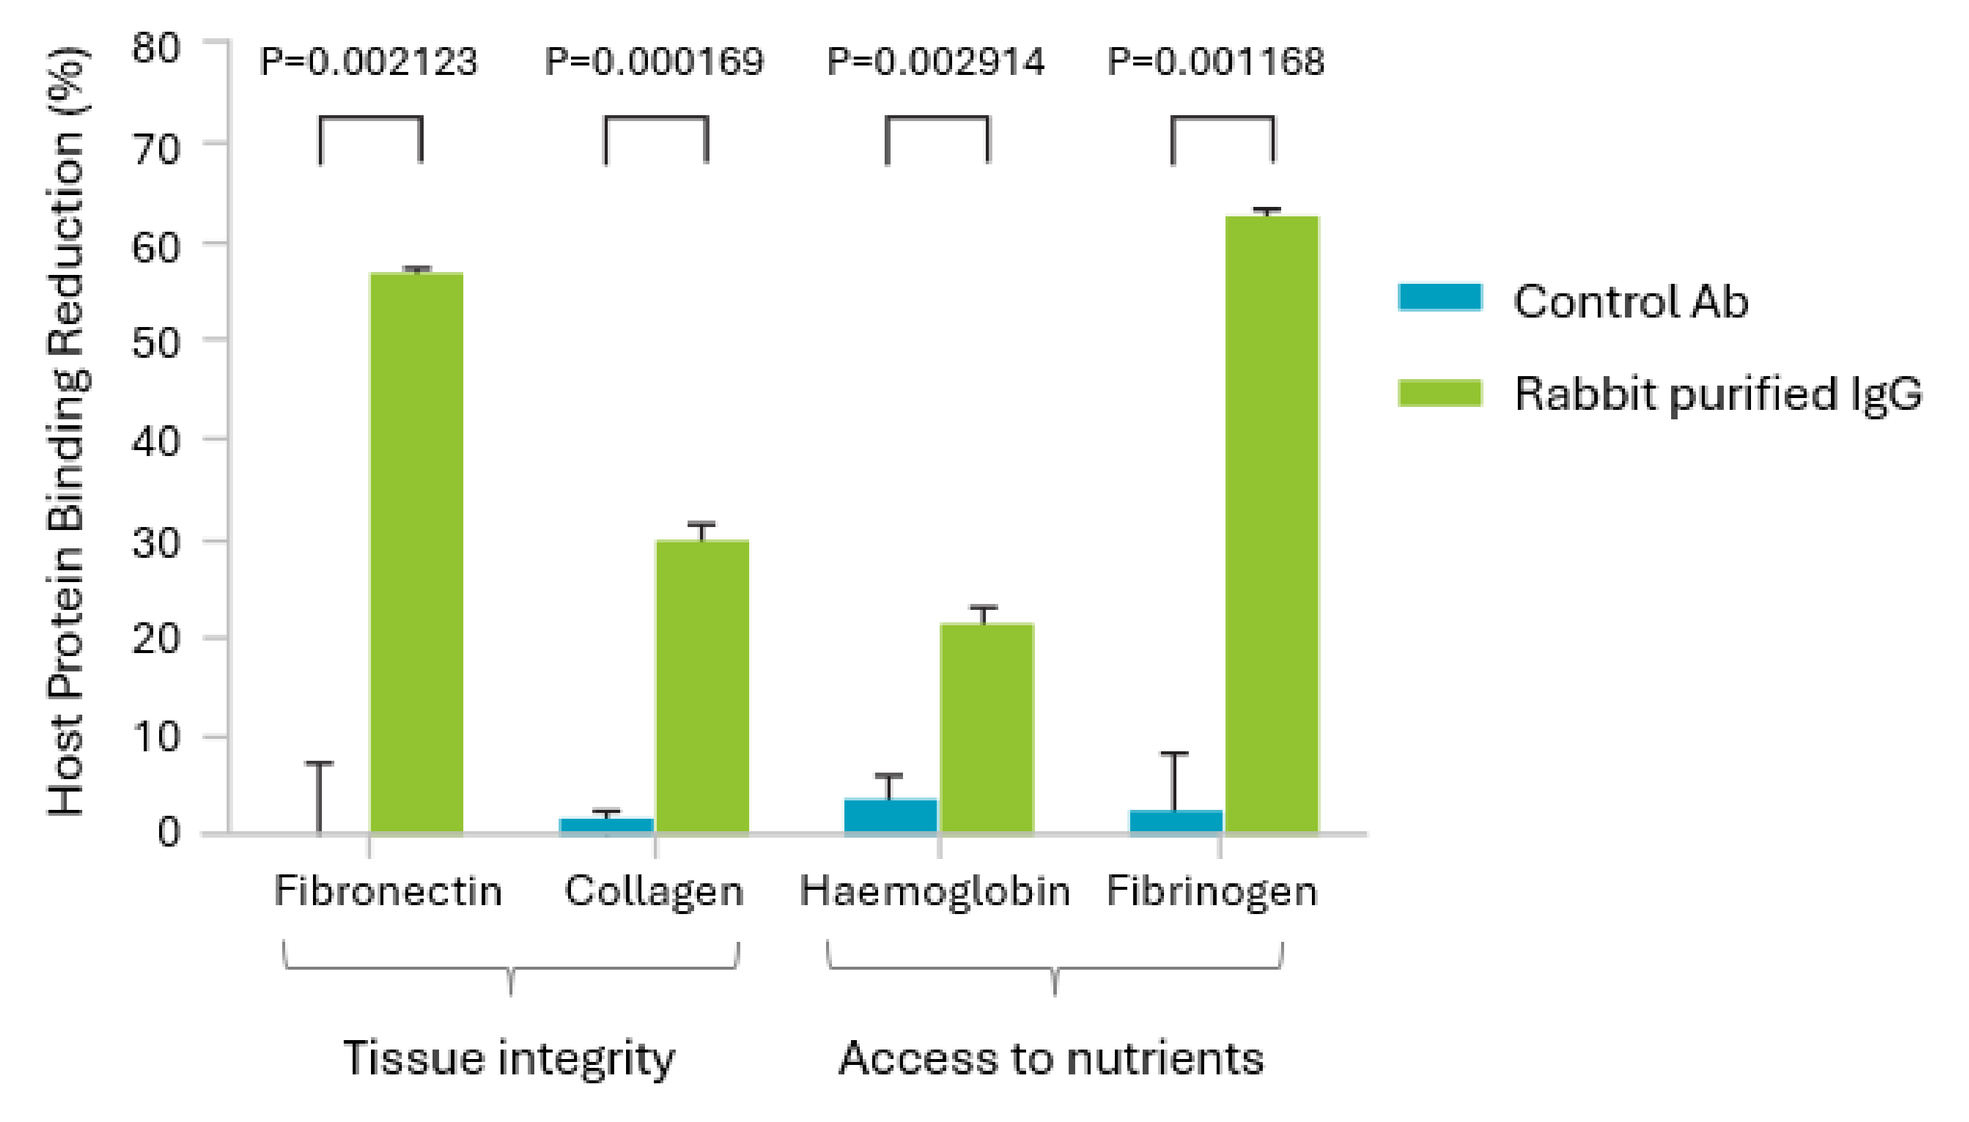
Host Protein Binding Reduction (%)
P=0.002123
P=0.000169
P=0.002914
P=0.001168
Control Ab
Rabbit purified IgG
Fibronectin
Collagen
Haemoglobin
Fibrinogen
Tissue integrity
Access to nutrients

Method: ELISA format – gingipains incubated with sera from vaccinated rabbit are inhibited from binding target antigen

Protease inhibition

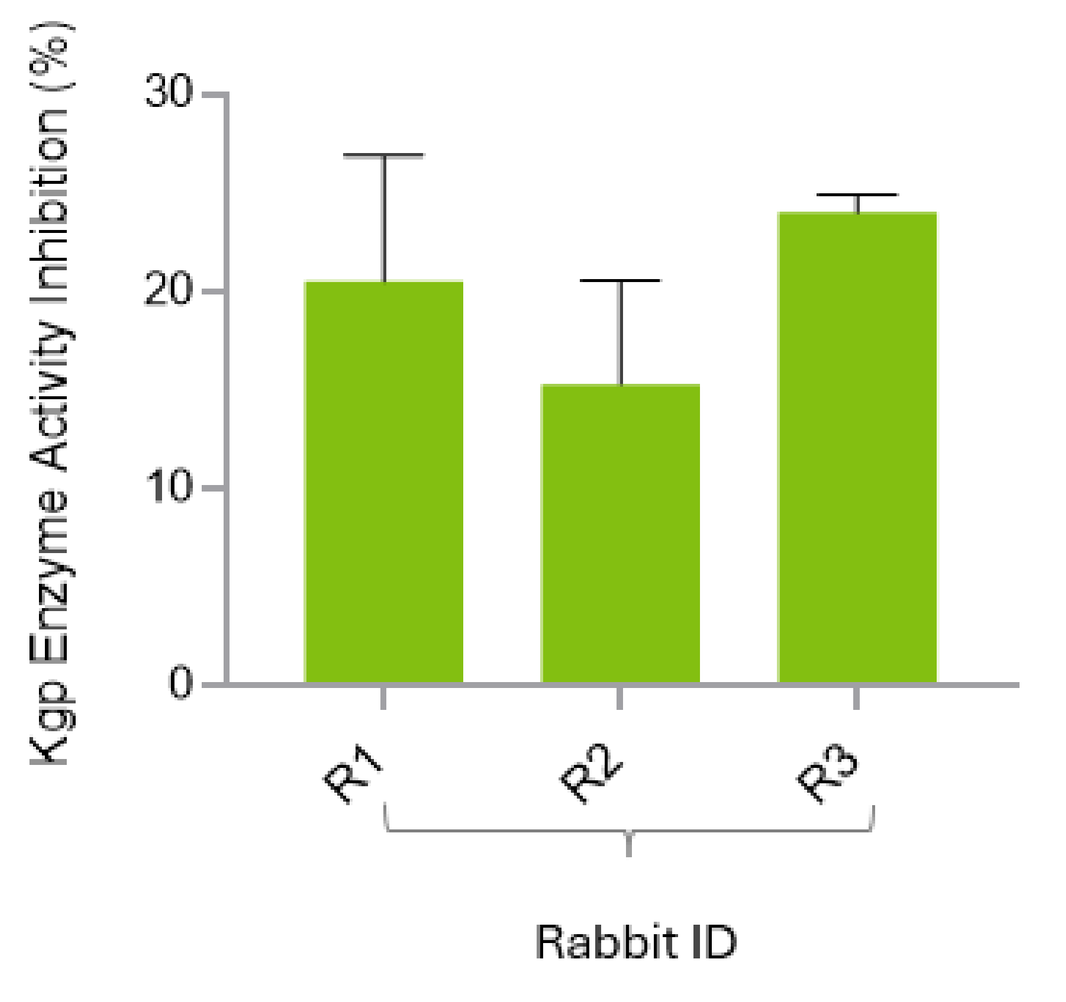
Kgp Enzyme Activity Inhibition (%)
R1
R2
R3
Rabbit ID

Method: Protease assay – gingipains incubated with sera from vaccinated rabbits are inhibited from cleaving chromogenic substrate

By neutralizing gingipain toxins, therapeutic dosing of GPV381 can limit bone loss in a challenge model

Dose-Dependent Protection Against Bone Loss

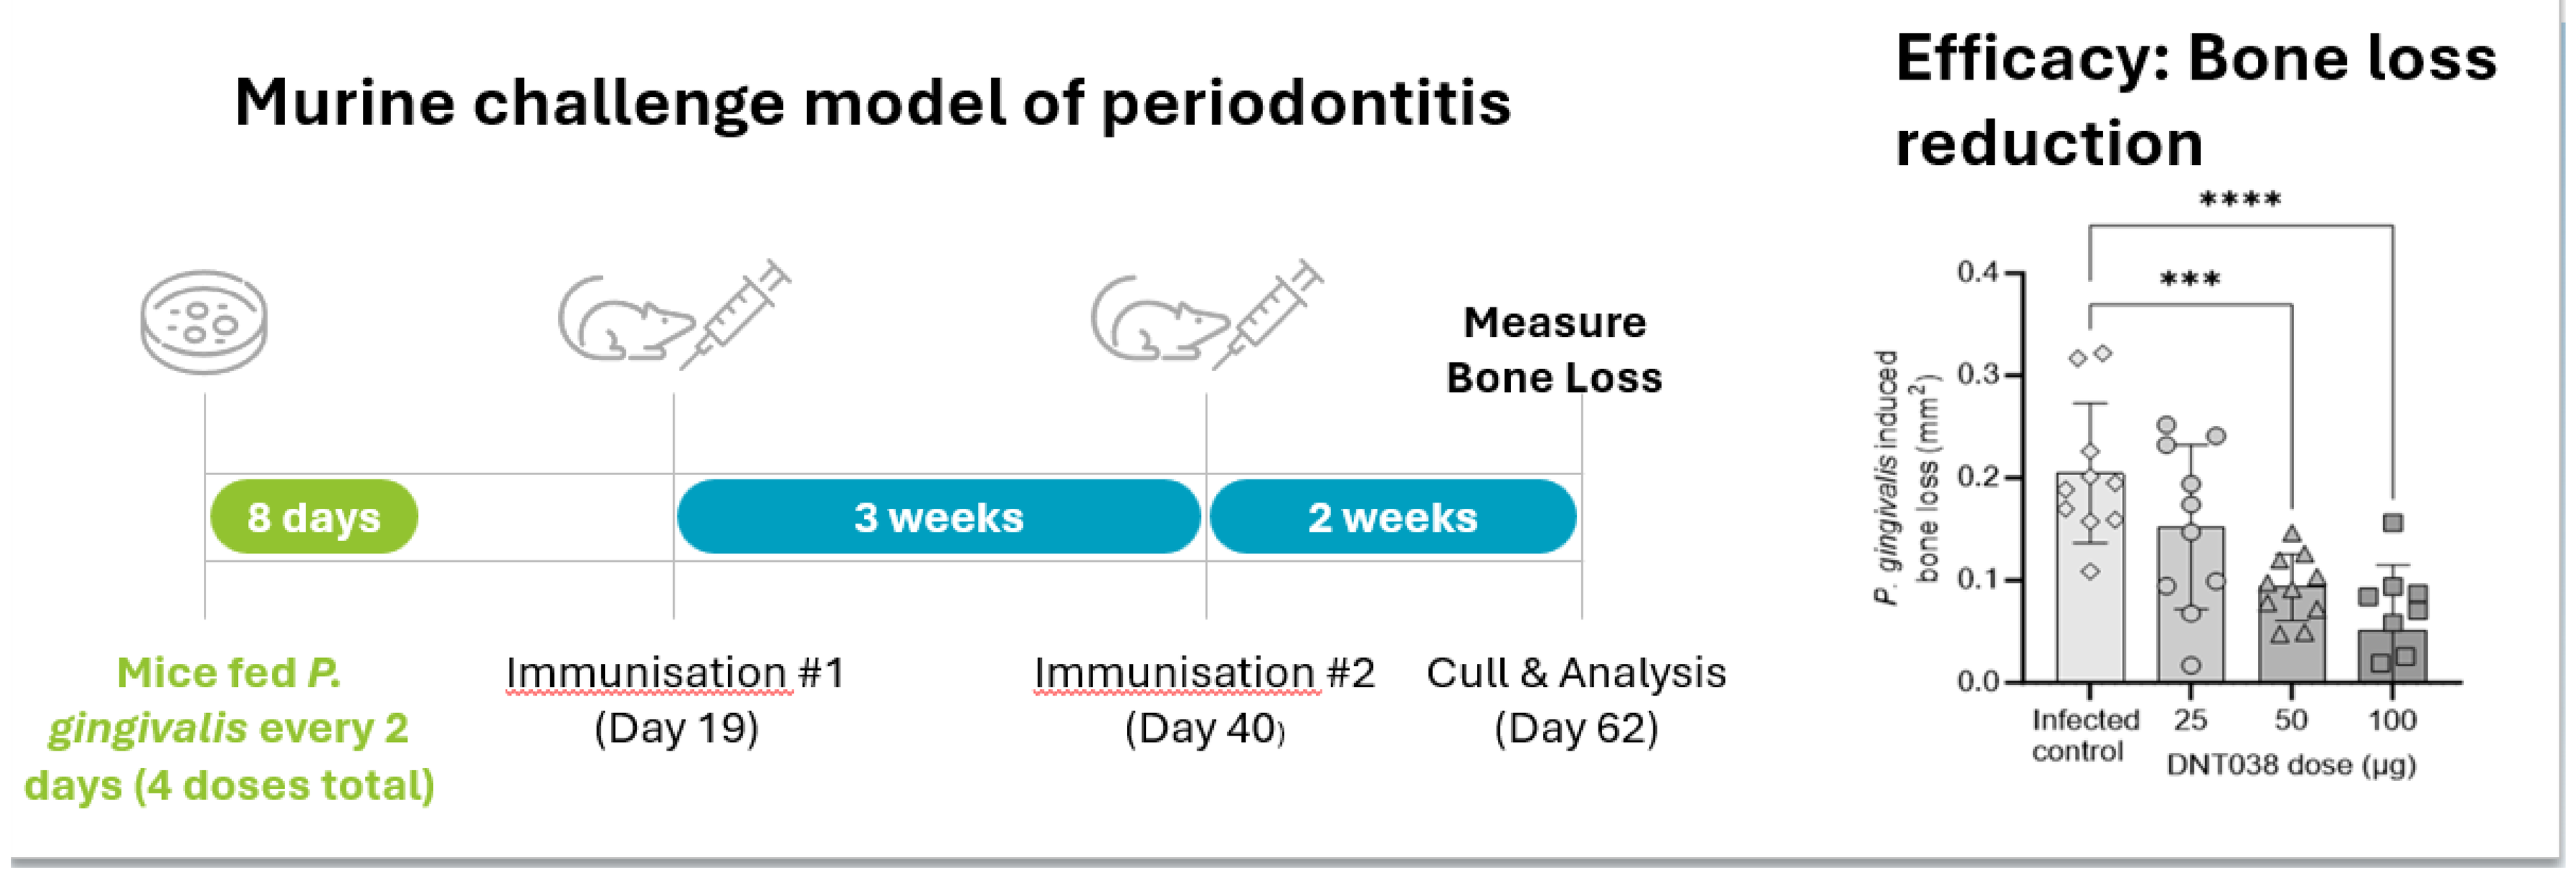
Murine challenge model of periodontitis
Measure Bone Loss
8 days
3 weeks
2 weeks
Mice fed P. gingivalis every 2 days (4 doses total)
Immunisation #1 (Day 19)
Immunisation #2 (Day 40)
Cull & Analysis (Day 62)
Efficacy: Bone loss reduction

P. gingivalis induced bone loss (mm²)
Infected control
25
50
100
DNT038 dose (µg)

Phase 1: Safe and well tolerated with durable, functional anti-gingipain antibodies for 12 months

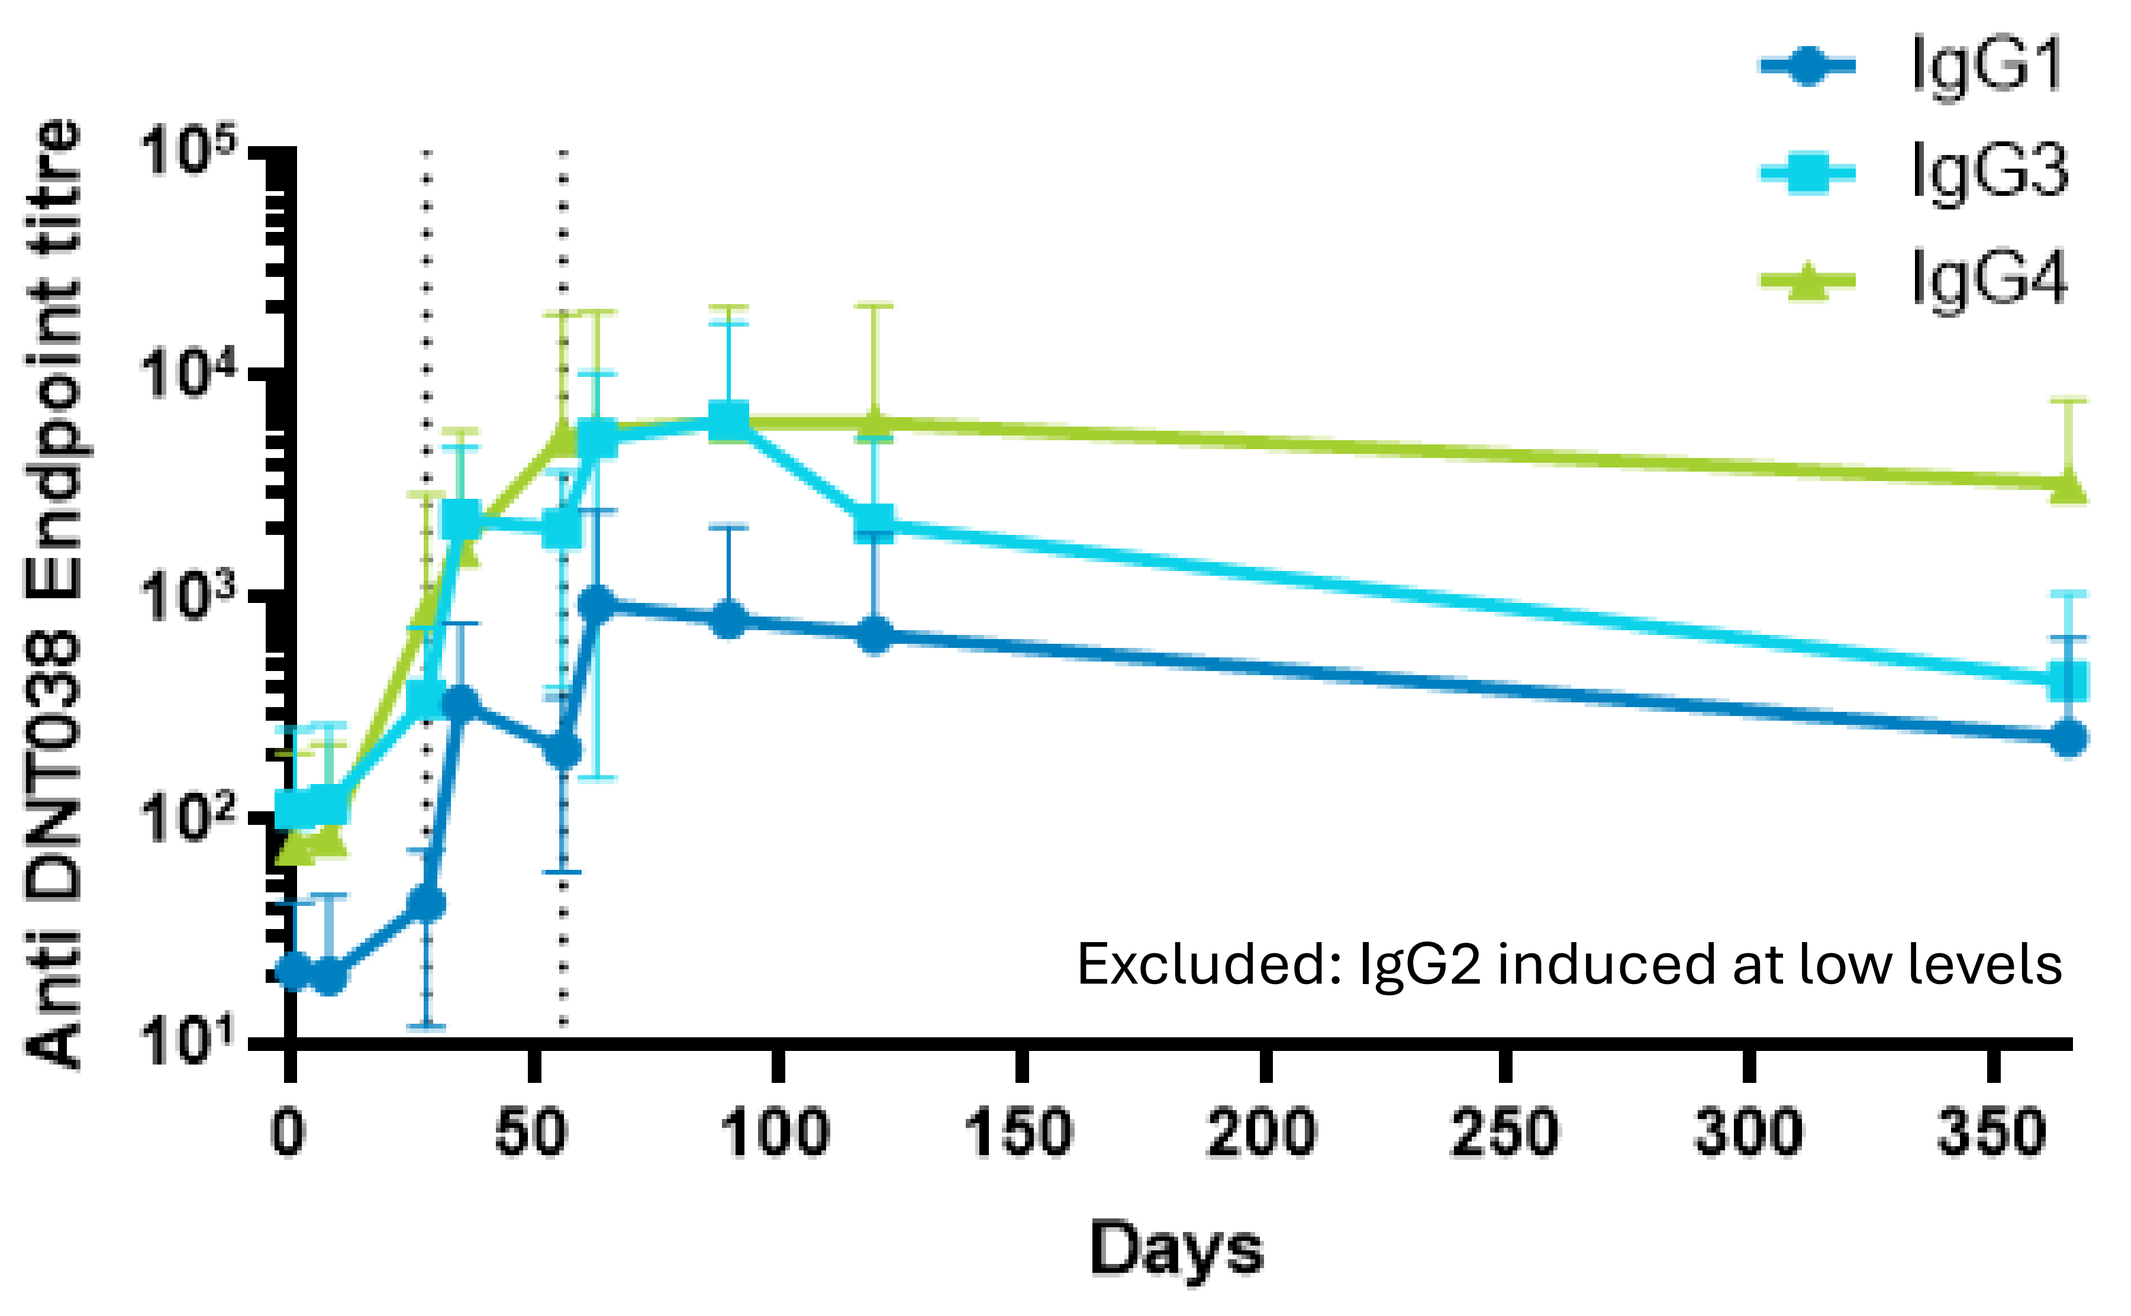
IgG1
IgG3
IgG4
Anti DNT038 Endpoint titre
Excluded: IgG2 induced at low levels
Days

- High titers persist for 12-months, especially IgG4
- These subclasses provide a range of functionality including the inhibition of inflammatory signaling

Neutralizing gingipains disrupts the pathogenic periodontal ecosystem

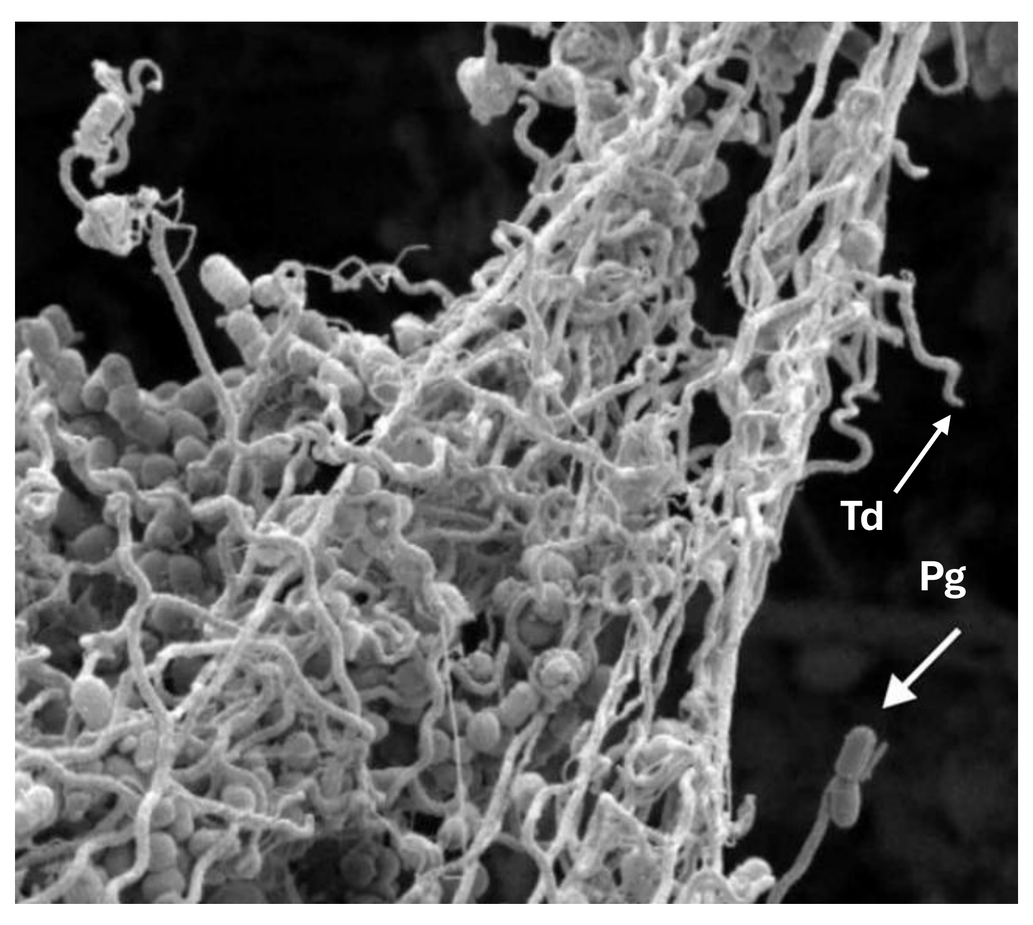
Td
Pg

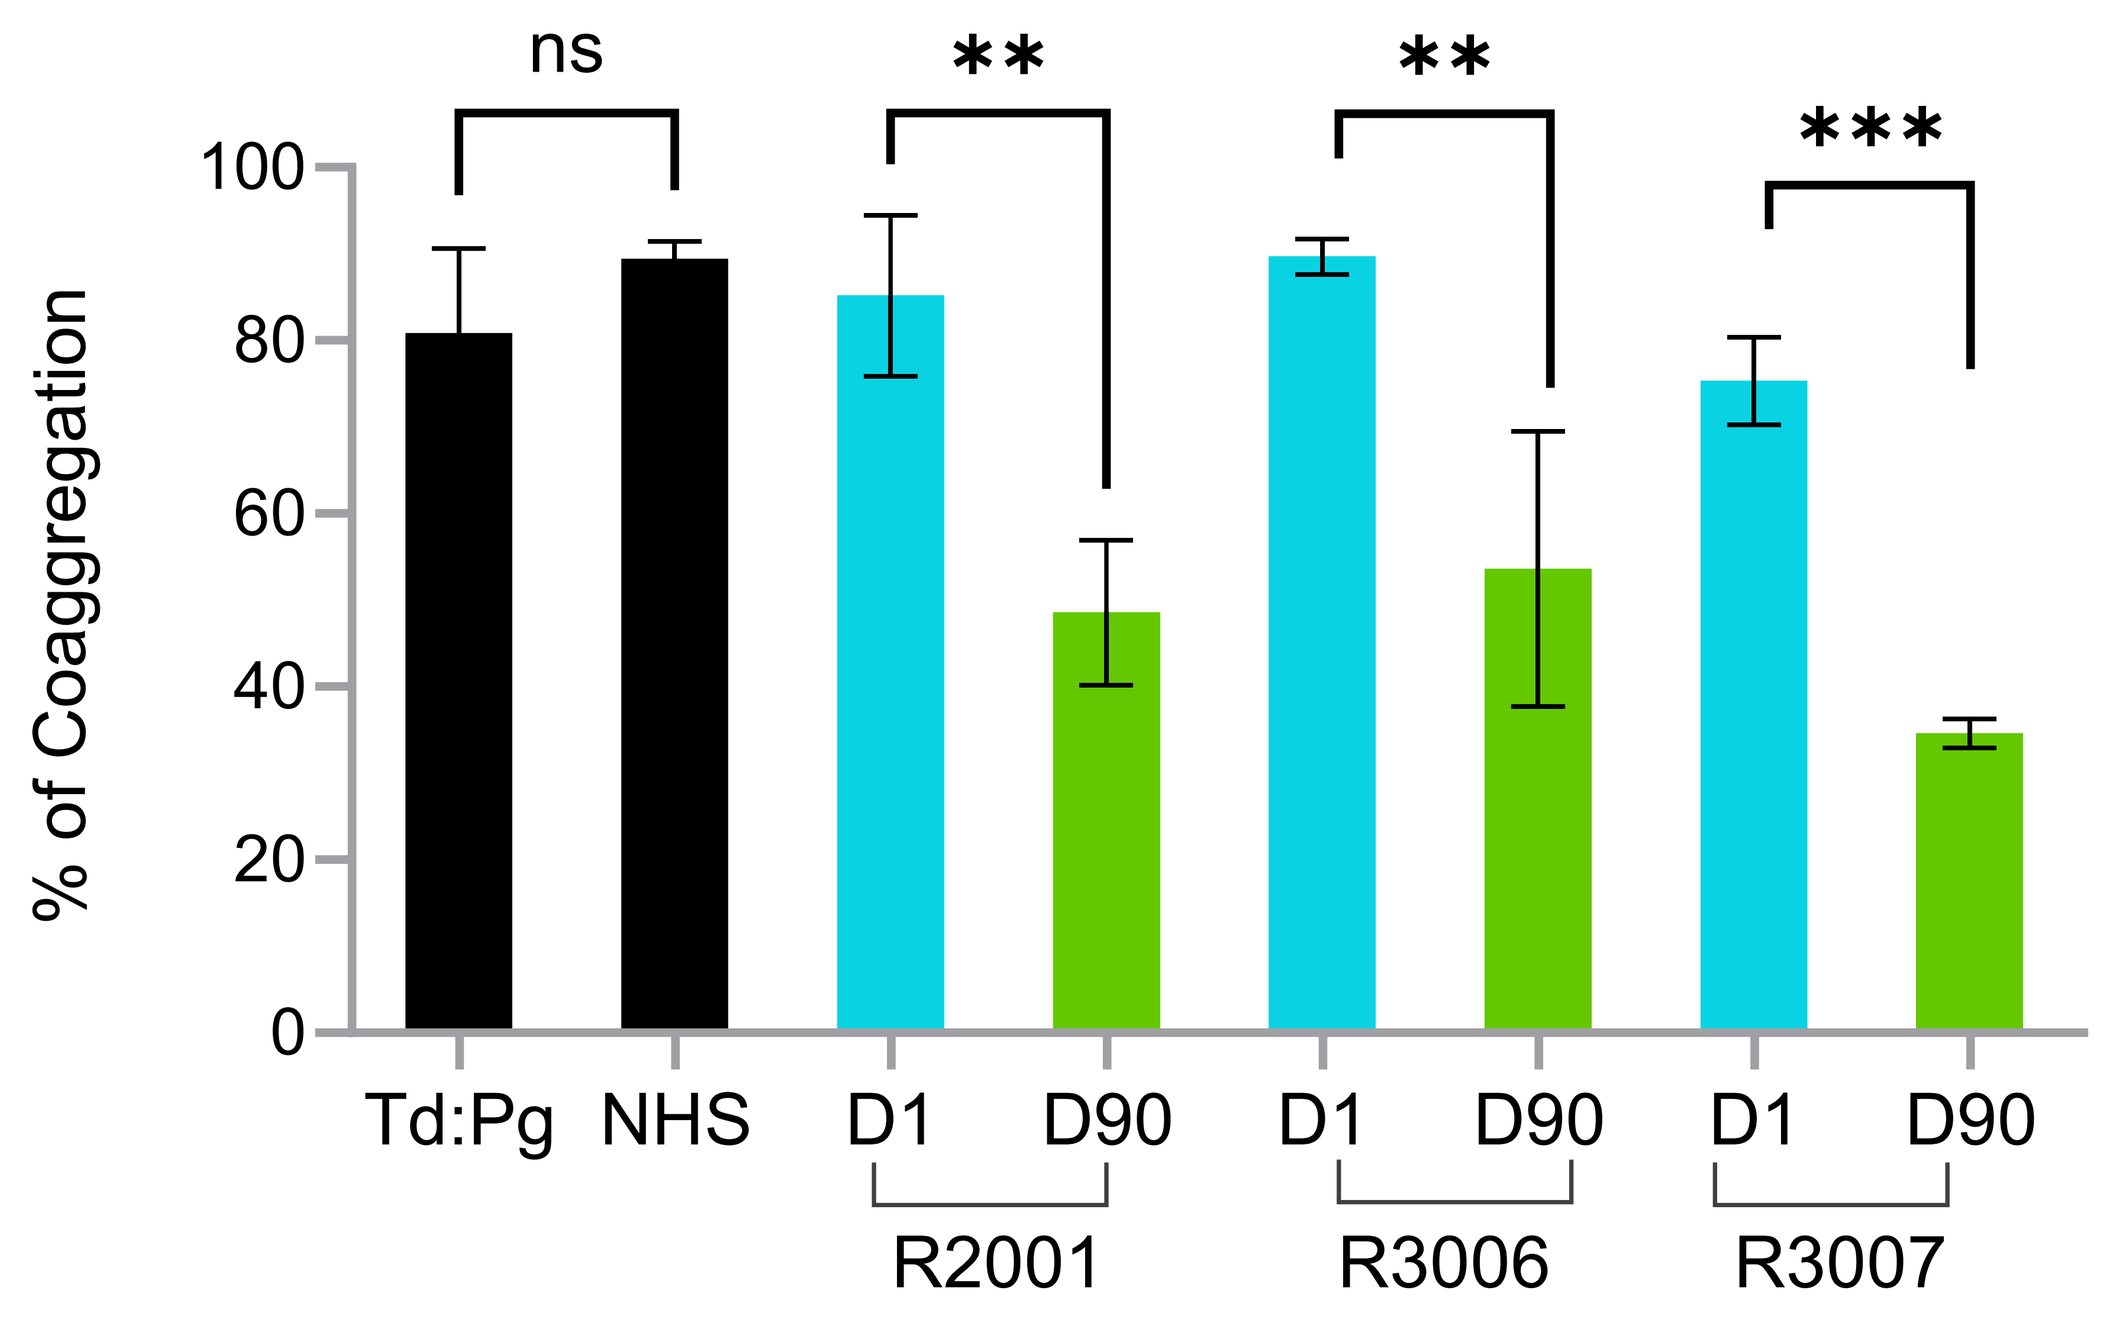
ns
**
**

% of Coaggregation
Td:Pg
NHS
D1
D90
R2001
R3006
R3007

- Surface gingipains enable coaggregation that enhances pathogenicity
- Ph1 Abs significantly inhibit coaggregation, collapsing the pathogenic ecosystem

REGAIN Phase II is designed to demonstrate disease modification in advanced periodontitis

REGAIN:

Restoration of symbiosis through **E**radication of **G**ingipain-**A**ssociated **In**flammation

A randomized, double blind, placebo-controlled study (n=174)

Population	Patients with advanced periodontitis (pockets ≥6 mm)
Arms	SRP + GPV381(Lo) vs. SRP + GPV381(Hi) vs. SRP + Placebo
Primary Objectives	Safety, dose finding, pocket depth reduction (primary efficacy endpoint)
Secondary Objective	Efficacy: Pocket closure and Bleeding on Probing (BoP); local and systemic inflammatory biomarkers
Doses	3 x intramuscular doses, administered 3 months apart
Trial Length	19 months: (3Q26 – 1Q28)

- GPV381 administered adjunct to SRP, with design similar to Arestin® clinical trial
- Primary endpoint: PPD reduction – FDA accepted primary endpoint
- Secondary endpoint: includes % Pocket closure (<4 or 5mm PPD and no BoP) demonstrating **healing** and **disease modification**

GPV381 has the potentiall to treat Periodontitis – a $154B gingipain-driven chronic inflammatory disease

- **Large Unmet Need:** Periodontitis is a $154B chronic inflammatory disease affecting ~90M Americans, with ~14M patients suffering severe disease
- **First-in-class:** GPV381 is a disease-modifying therapeutic vaccine designed to neutralize gingipain toxins, upstream drivers of immune dysfunction and chronic inflammation
- **Clinically De-risked:** Phase 1 data demonstrate safety and durable neutralizing antibodies against gingipain toxins lasting 12 months.
- **Next Inflection Points:** IND Accepted, Phase IIb study planned for mid-2026, supported by a $40–50 M raise to demonstrate clinical efficacy
- **Pipeline in a Product Potential:** Gingipain-driven inflammation is implicated in multiple systemic conditions including cardiovascular disease, diabetes, and Alzheimer's disease.